Jean-Bernard Gruenberger; jean-bernard.gruenberger@novartis.com

Poster xxxx

An international, longitudinal, observational study to evaluate the psychometric properties of a daily diary for adolescents and adults with sickle cell disease

Avery A. Rizio[1], Michelle K. White[1], Lindsay Petrenchik[2], Anne Ruault[2], Jean-Bernard Gruenberger[2], Kwaku Marfo[2], Kenneth I. Ataga[3]

[1]QualityMetric Incorporated LLC, Johnston, RI, USA; [2]Novartis Pharma AG, Basel, Switzerland; [3]UTHSC Center for Sickle Cell Disease, Memphis, TN, USA

This study is sponsored by Novartis Pharma AG
Poster presented at the ASCAT-EHA-BSH Sickle Cell Disease Conference 2022, held in London, United Kingdom on 20–22 October 2022

Scan to obtain:
- Poster
- Plain language summary
- Slides
- Supplementary material

http://novartis.medicalcongressposters.com/Default.aspx?doc=xxxxx

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- **Primary Conclusion**: The SCPD-S Daily VOC Impact score, comprised of items 11-16, is a reliable and valid measure of VOC impact, and is suitable for use in trials, observational studies, and clinic settings
- The SCPD-S evaluates VOC frequency and duration, experiences during a VOC (e.g., medication use, healthcare visits), and provides a daily measure of VOC impact
- Compliance with completing the daily diary was 87%
- The SCPD-S Daily VOC Impact Scale 1 (items 11-16) showed sufficient evidence of reliability and validity across psychometric properties. Scale 2 had weaker evidence, particularly in its ability to detect change
 - Scale 1 should be used as a measure of daily VOC impact
 - Scale 2 should be dropped from the diary
- Limitations: difficulty identifying stable patients for evaluation of test-retest reliability due to the inherent variability in VOCs, use of a convenience sample, and data collection during the COVID-19 pandemic, which could impact experiences during a VOC
- Strengths: use of a study design that supports the feasibility of the daily diary in patients across ages and geographic regions, and use of development and validation procedures that followed best practice guidelines

INTRODUCTION

- Vaso-occlusive crises (VOCs) are caused by multi-cell adhesion or cell clusters that block or reduce blood flow, and are one of the most serious complications of sickle cell disease (SCD)[1-3]
- Because VOCs occur sporadically and unpredictably, a daily diary – the Sickle Cell Pain Diary - Self Report (SCPD-S) – was developed to more accurately capture the frequency, duration, severity of pain, and related impacts experienced during a VOC, including VOCs managed at home (**Figure 1**)[4]
 - Each day, respondents indicate if they have experienced a VOC (referred to as a “sickle cell pain crisis”) in the past 24 hours
 - If they have not experienced a VOC, respondents answer 5 questions on the impact of SCD over the past 24 hours
 - If they have experienced a VOC, respondents answer 19 questions about the VOC over the past 24 hours

OBJECTIVE

To evaluate the measurement model, scoring, and psychometric properties of the SCPD-S VOC impact through data collected from an international, longitudinal, observational study

METHODS

Participant eligibility:
- Confirmed diagnosis of SCD
- Age 12 or older
- Currently live in the United States (US), United Kingdom (UK), or Kingdom of Saudi Arabia (KSA)
- Experienced ≥1 VOC in the past 12 months

Participants were recruited through participant panels, advocacy groups, and clinician referrals

Study Design:
- The survey was administered through a mobile application downloaded onto patients’ devices
- Patients completed survey questions every day for at least 3 months
 - The SCPD-S was administered every day
 - Other assessments were administered at pre-specified intervals, which served as criterion variables for planned analyses (**Figure 2**)
- Patients could backfill up to 2 days of missed surveys
- The analytic sample included patients who experienced at least 1 VOC while enrolled in the study and who completed ≥25 of 30 daily assessments during the month the VOC occurred

Measurement Model:
- Multi-level factor analyses were conducted in a nested setting, where daily assessments were nested within each individual patient (within- and between-patient levels)
- The hypothesized unidimensional model of VOC impact was tested with a confirmatory factor analysis (CFA); split-half exploratory factor analyses (EFAs) and follow-up CFAs were conducted to explore alternative/refined models
 - Acceptable model fit: confirmatory fit index (CFI) ≥0.90; root mean square error of approximation (RMSEA) ≤0.08; standardized root mean square residual (SRMR) ≤0.08
- Cronbach’s alpha evaluated internal consistency reliability
 - Acceptable Cronbach’s alpha: 0.70

Item-Level Properties:
- Analyses of item-level data, including evaluation of item/floor effects, Mokken scale analyses, and inter-item correlations were conducted on VOC impact items. Results were satisfactory and are not presented here

Differential Item Functioning (DIF):
- Analyses examined whether different groups of patients respond differently to VOC impact items, even after controlling for levels of the underlying latent trait
- DIF due to age and country of residence was examined using a series of ordinal logistic regression models

SCPD-S Scoring:
- Three different SCPD-S VOC impact scores were investigated:
 - SCPD-S Daily VOC Impact Score: the overall impact of the VOC on a given day. One score is produced for each day a VOC is experienced
 - SCPD-S 7-Day VOC Impact Aggregate: the average of daily scores over a 7-day period
 - VOC Experience Impact Aggregate: the average of daily scores over days on which a VOC was experienced
- All scores range from 0-100; higher scores indicate more impact
- This poster focuses on psychometric analyses of the Daily VOC Impact Score

Psychometric Analyses:
- **Test-Retest Reliability:** Intraclass correlation coefficients (ICC) evaluated agreement between scores at different time points among a subset of stable patients defined by responses on the Patient Global Impression of Severity (PGI-S) item
 - Moderate ICC: ≥0.50; Good ICC: ≥0.75
- **Convergent Validity:** Pearson correlations were conducted between VOC impact scores and criterion variables
 - Acceptable convergent validity: ≥0.30
- **Known-Groups Validity:** VOC impact scores were compared across groups of patients who differ according to pre-defined criteria
 - Acceptability interpreted through patterns of means, effect sizes (ES), and p-values
 - ES interpretation: Negligible: 0.2< |ES|; Small: 0.2≤ |ES| <0.5; Medium: 0.5≤ |ES| <0.8; Large: |ES| ≥0.8
- **Responsiveness:** Change in VOC impact scores were correlated with change in criterion variables. Mean change in VOC impact scores were also examined as a function of change in symptom severity
 - Acceptable correlations: ≥0.30; mean change: interpreted through patterns of means, ES, and p-values

Figure 1. SCPD-S Item Content

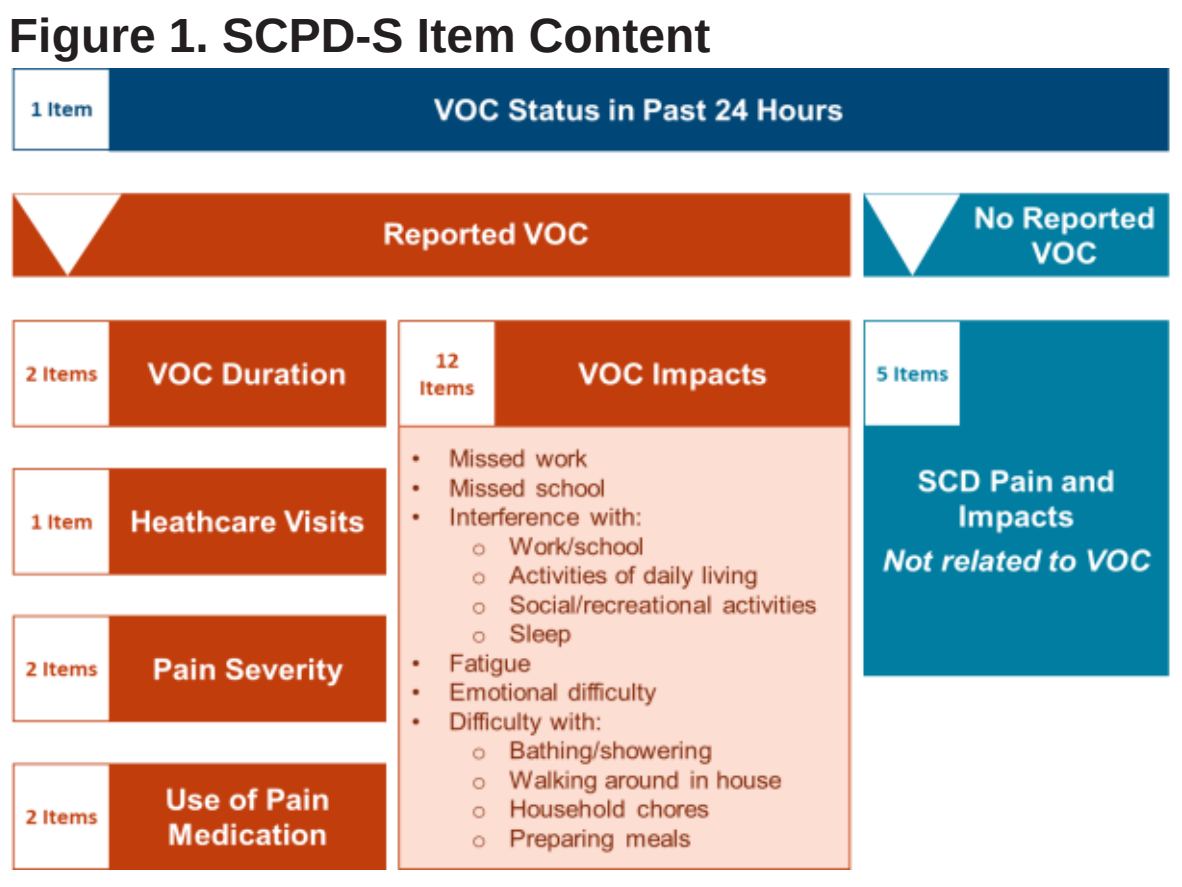
1 Item
VOC Status in Past 24 Hours
Reported VOC
No Reported VOC
2 Items
VOC Duration
12 Items
VOC Impacts
5 Items
SCD Pain and Impacts
Not related to VOC
1 Item
Heathcare Visits
Missed work
Missed school
Interference with:
Work/school
Activities of daily living
Social/recreational activities
Sleep
Fatigue
Emotional difficulty
Difficulty with:
Bathing/showering
Walking around in house
Household chores
Preparing meals
2 Items
Pain Severity
2 Items
Use of Pain Medication

Figure 2. Study Assessment Schedule

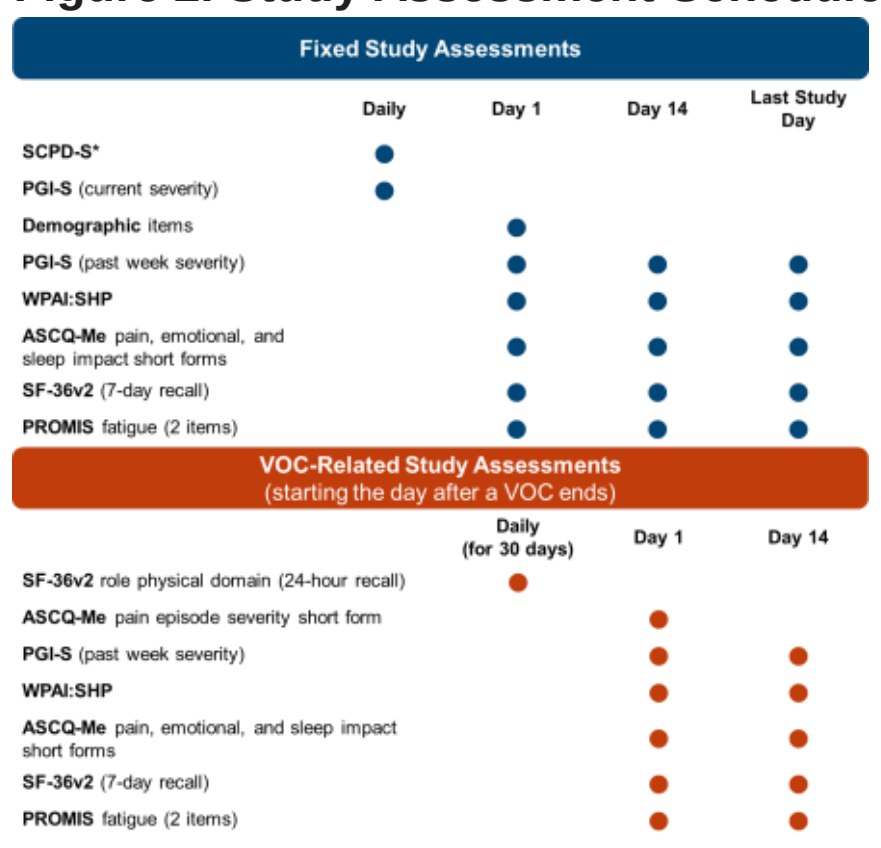
Fixed Study Assessments
Daily
Day 1
Day 14
Last Study Day
SCPD-S*
PGI-S (current severity)
Demographic items
PGI-S (past week severity)
WPAI:SHP
ASCQ-Me pain, emotional, and sleep impact short forms
SF-36v2 (7-day recall)
PROMIS fatigue (2 items)
VOC-Related Study Assessments
(starting the day after a VOC ends)
Daily (for 30 days)
Day 1
Day 14
SF-36v2 role physical domain (24-hour recall)
ASCQ-Me pain episode severity short form
PGI-S (past week severity)
WPAI:SHP
ASCQ-Me pain, emotional, and sleep impact short forms
SF-36v2 (7-day recall)
PROMIS fatigue (2 items)

RESULTS

Patient Sample

- The analytic sample included 299 patients (**Figure 3**)
- Demographic characteristics of the analytic sample and experiences while enrolled in the study are presented in **Table 1**

Figure 3. Overview of Patient Disposition

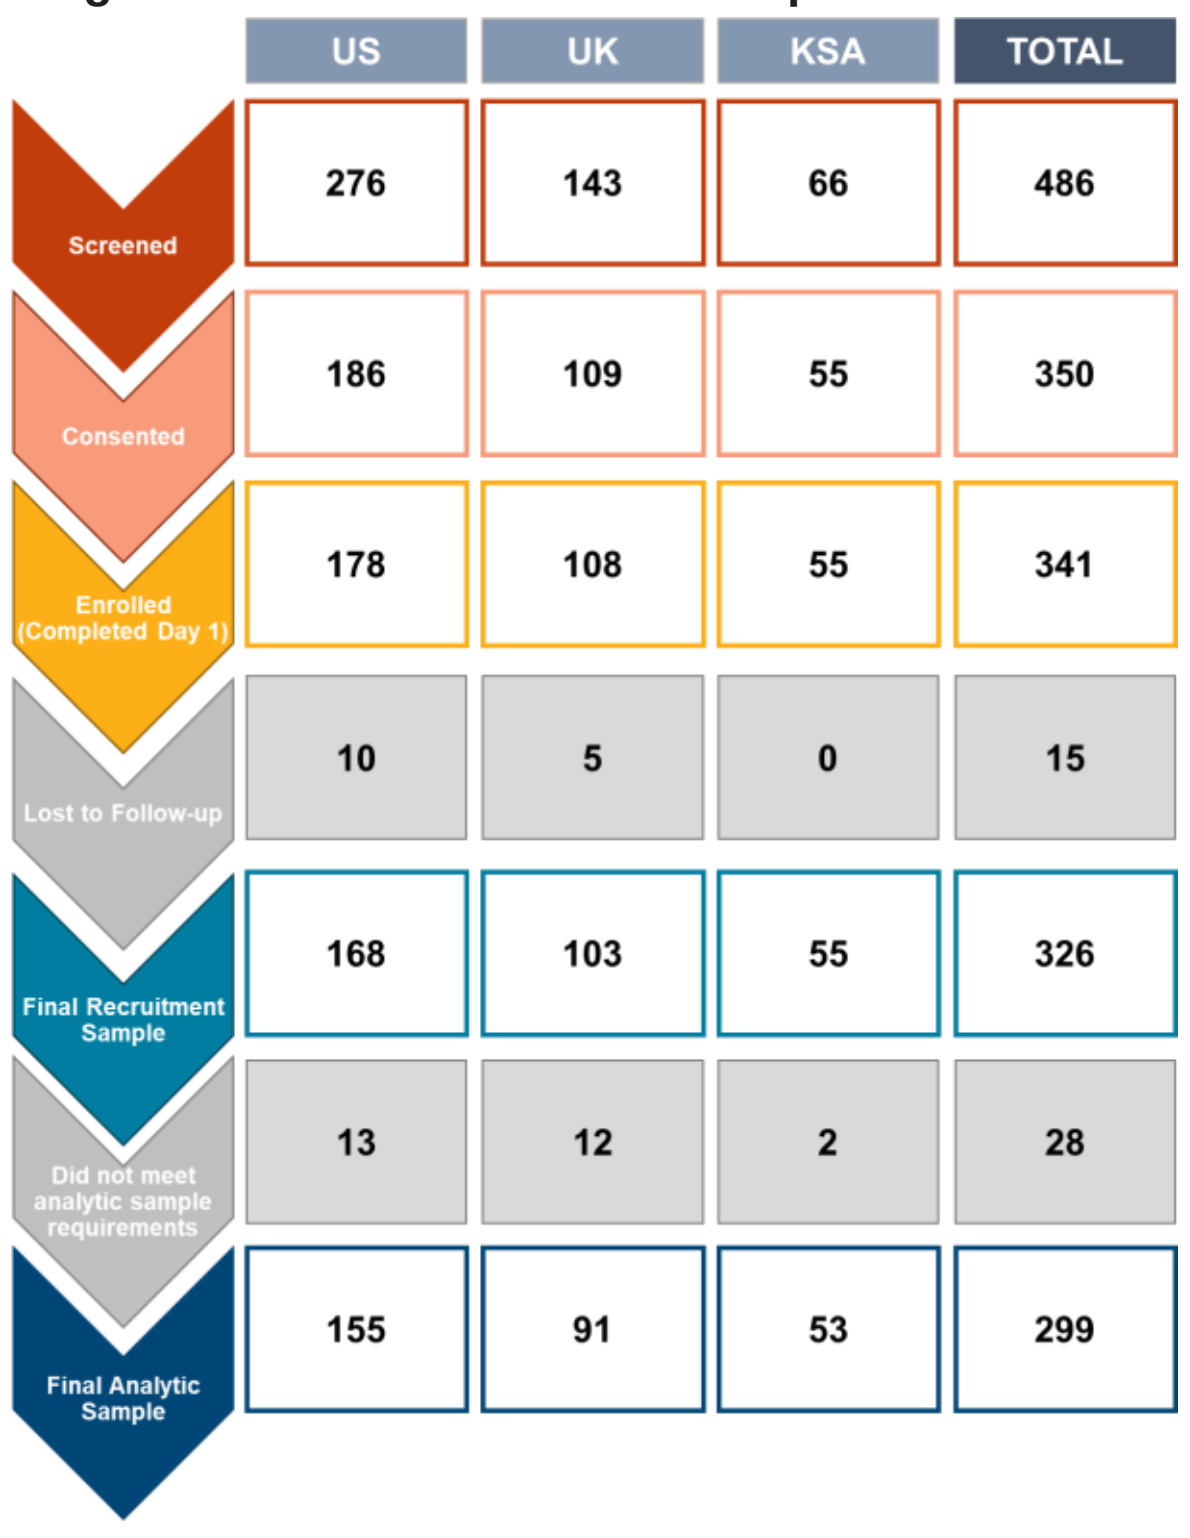
US
UK
KSA
TOTAL
Screened
276
143
66
486
Consented
186
109
55
350
Enrolled (Completed Day 1)
178
108
55
341
Lost to Follow-up
10
5
0
15
Final Recruitment Sample
168
103
55
326
Did not meet analytic sample requirements
13
12
2
28
Final Analytic Sample
155
91
53
299

Table 1. Patient Characteristics

Demographics	Mean (SD) / N (%)
Age, years	30.3 (10.24)
Gender, female	215 (72%)
Race, Black or African	246 (83%)
Employment Status	
Student	79 (27%)
Employed (full or part time)	85 (29%)
Disabled/unable to work	64 (22%)
Unemployed	42 (14%)
Other	28 (9%)
SCD Type	
Hb-SS	159 (53%)
Hb-SC	55 (19%)
Hb-S beta thalassemia (+ or 0)	29 (10%)
Hb-SD	1 (0.3%)
Hb-SO	1 (0.3%)
Not known	53 (18%)
Experiences in the study	**Mean (SD)**
Number of VOCs experienced	6.8 (5.35)
Duration of first VOC, days	7.0 (19.19)
Duration of study enrollment, days	100.8 (18.65)

Of the 299 patients in the study, 1 patient is missing demographic information.

Measurement Model

Factor Analyses

- The unidimensional model with constrained factor loadings yielded poor fit (**Table 2, Model 1**); a model with unconstrained factor loadings showed nearly identical results (data not shown)
- Two EFAs examined model fit for a 2-factor (**Table 2, Model 2**) and 3-factor solution (data not shown)
 - The 2-factor solution was selected, as it was the simplest model with good fit
- A 2-factor CFA with constrained factor loadings was conducted (**Table 2, Model 3**)
 - Refinements to the model were considered, allowing for unconstrained factor loadings and for a single item to cross-load onto both factors (data not shown)
 - The model with constrained factor loadings across levels was retained as the final model for parsimony (i.e., Model 3). Item content for each factor is shown in **Figure 4**
- A bi-factor model indicated that the measurement model is not suited to a single general factor (data not shown)

Table 2. Model Fit Statistics

Fit Statistics	Model 1	Model 2	Model 3
χ^2	2034.57 (df=79)	307.18 (df=52)	286.45 (df=76)
CFI	0.78	0.95	0.95
RMSEA	0.11	0.07	0.06
SRMR			
Within	0.10	0.04	0.03
Between	0.10	0.05	0.07

Model 1: Unidimensional CFA; Model 2: 2-Factor EFA; Model 3: 2-Factor CFA with constrained factor loadings (final model).

Figure 4. Two-Factor Model of SCPD-S VOC Impact

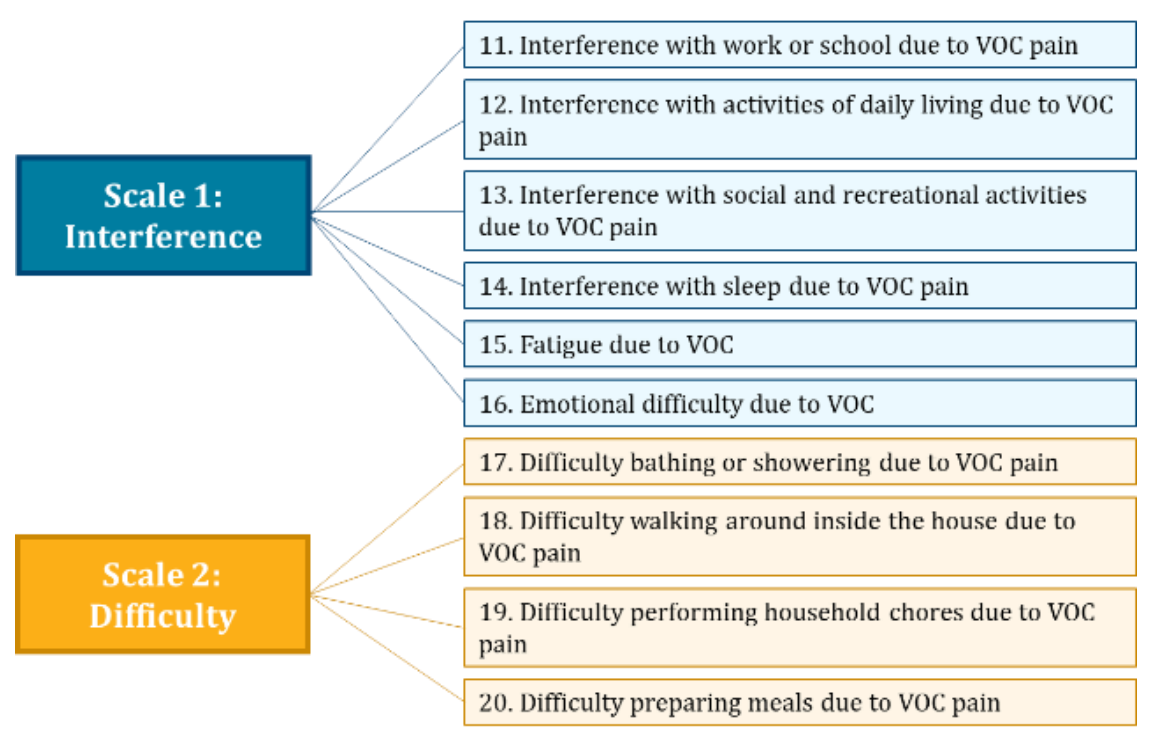
Scale 1: Interference
Scale 2: Difficulty
11. Interference with work or school due to VOC pain
12. Interference with activities of daily living due to VOC pain
13. Interference with social and recreational activities due to VOC pain
14. Interference with sleep due to VOC pain
15. Fatigue due to VOC
16. Emotional difficulty due to VOC
17. Difficulty bathing or showering due to VOC pain
18. Difficulty walking around inside the house due to VOC pain
19. Difficulty performing household chores due to VOC pain
20. Difficulty preparing meals due to VOC pain

Internal Consistency Reliability

- Internal Consistency Reliability of the two factors was acceptable
 - Factor 1, Cronbach’s alpha = 0.87
 - Factor 2, Cronbach’s alpha = 0.86

Differential Item Functioning

- Neither age nor country of residence substantially increased the amount of explained variance in item responses to the SCPD-S
- Results show that when conditioning on the latent trait, respondents of different ages and different countries of residence endorse SCPD-S VOC impact items in a similar manner
- DIF due to age and country of residence is not a concern for Scale 1 or Scale 2 SCPD-S items

Psychometric Properties of the SCPD-S Daily VOC Impact Score

Test-Retest Reliability

- Test-retest reliability was good for Scale 1 (N = 114, ICC = 0.78 [95% confidence interval = 0.71-0.86]) and moderate for Scale 2 (N = 114, ICC = 0.66 [95% confidence interval = 0.56-0.76])

Convergent Validity

- Both scales were correlated with VOC pain severity at its worst and at its least (**Table 3**)
- Scale 1 showed slightly stronger relationships with pain severity than Scale 2

Known-Groups Validity

- Patients with greater symptom severity had significantly higher SCPD-S Daily VOC Impact scores (indicating greater impact) than patients with lower symptom severity, as defined by the PGI-S (p<0.001; **Table 4**)
- Effect sizes used to interpret the magnitude of difference in the daily scores are considered large (≥0.80)

Responsiveness

Correlation analyses
- Change in SCPD-S Daily VOC Impact scores for Scale 1 were correlated with change in symptom severity (r=0.41); the strength of these relationships exceeded the threshold considered indicative of adequate responsiveness
- Change in Scale 2 scores were not as strongly correlated with change in symptom severity (r=0.27)

Mean Change Analyses (**Table 5**)
- For both scales, significant differences in mean SCPD-S Daily VOC Impact change scores were observed across groups of patients defined by change in symptom severity (p<0.001). The greatest amount of improvement in SCPD-S scores was observed for patients whose change in PGI-S indicated improvement

Table 3. Convergent Validity of SCPD-S Daily VOC Impact Score

Criterion measure	Scale 1 Coefficient	Scale 2 Coefficient
VOC pain at its worst	0.49	0.44
VOC paint at its least	0.44	0.36

Scale 1 N = 299; Scale 2 N = 296.

Table 4. Known-Groups Validity of the SCPD-S Daily VOC Impact Score

	Scale 1		Scale 2	
	N	Mean (SD)	N	Mean (SD)
PGI-S Group[1]				
Higher symptom severity	90	74.2 (18.94)	90	63.1 (25.85)
Lower symptom severity	209	48.4 (23.98)	206	36.4 (28.55)
ES	1.15		0.97	
F (P-value)	82.01 (P<.001)		58.16 (P<.001)	

[1] Higher symptom severity: PGI-S = Extremely, very, or markedly severe; Lower symptom severity: PGI-S = Not at all, minimally, mildly, or moderately severe.

Table 5. Responsiveness of the SCPD-S Daily VOC Impact Score, Mean Change

	Scale 1			Scale 2		
	N	Mean Change (SD)	ES	N	Mean Change (SD)	ES
Change in PGI-S						
Improved	58	-16.5 (21.04)	-0.75	58	-14.2 (23.25)	-0.46
No Change	59	-9.4 (15.83)	-0.42	59	-7.1 (25.17)	-0.25
Worsened	46	9.2 (19.62)	0.40	44	14.2 (29.46)	0.50
F (P-value)	24.7 (P<0.001)			15.9 (P<0.001)		

Patients were grouped according to change in between the first day and the middle day of the first VOC. Since higher SCPD-S scores indicate greater impact, a negative change score indicates improvement (i.e., reduced impact), while a positive change score indicates worsening (i.e., increased impact).

Psychometric Properties of the SCPD-S 7-Day VOC Impact and VOC Experience Impact Aggregate Scores

- Results of psychometric analyses of the SCPD-S 7 Day VOC Impact Aggregate and SCPD-S VOC Experience Impact Aggregate scores are summarized in **Table 6**
 - Scale 1, but not Scale 2, of the VOC Experience Impact Aggregate score showed adequate psychometric properties
 - Psychometric properties for both scales of the 7-Day VOC Impact Aggregate were not satisfactory

Table 6. Summary of Psychometric Analyses of the SCPD-S VOC Impact Aggregate Scores

	SCPD-S 7 Day VOC Impact Aggregate		SCPD-S VOC Experience Impact Aggregate	
	Scale 1	Scale 2	Scale 1	Scale 2
Test-retest reliability	Unsatisfactory	Unsatisfactory	Satisfactory	Satisfactory
Convergent validity	Satisfactory	Satisfactory	Satisfactory	Satisfactory; notably weaker than Scale 1
Known-groups validity	Borderline	Borderline	Satisfactory	Borderline; notably weaker than Scale 1
Responsiveness	Borderline	Unsatisfactory	Borderline	Borderline

Acknowledgements

We would like to thank the patients who participated in this study; their time, interest, and insight is greatly appreciated.

Disclosures

This study was sponsored by Novartis Pharma AG. LP, AR, JBG, and KM are employees of Novartis. AAR and MKW are employees of QualityMetric Incorporated, LLC, and received funding from Novartis to conduct this research. KIA has received honoraria from Global Blood Therapeutics, Editas Medicine, Novartis, Novo Nordisk, Modus Therapeutics, and Bioverativ; membership on an entity’s Board of Directors or advisory committees for Novartis, Global Blood Therapeutics, Novo Nordisk, Forma Therapeutics, and Roche; and research funding from Global Blood Therapeutics, Shire/Takeda, and Novartis.

References

1. Kato GJ, Piel FB, Reid CD, et al. Nat Rev Dis Primers. 2018;4:18010. 2. Piel FB, Steinberg MH, Rees DC. N Engl J Med. 2017;376(16). 3. Kanter J, Kruse-Jarres R. Blood Rev. 2013;27(6):279-287. 4. White MK, Saucier C, Bailey M, et al. J Patient Rep Outcomes. 2021;5(1):63.